Abstract ID 14867

AU-007, a computationally designed human antibody, mediates human immune activation by endogenous IL-2, while uniquely breaking the IL-2 negative feedback loop and preventing Treg expansion

Inbar Amit, Itay Levin*, Timothy Wyant*‡, Natali Levitin*, May Ben-Mayor*, Olga Bluvshtein*, Noam Grossman*, Reut Barak*, Guy Nimrod*, Michael Zehnin*, Sharon Fischman*, Marek Štrajbl*, Yehezkel Sasson*, Liron Danielpur*, James Vasselli‡, Aron Knickerbocker‡, Yanay Ofran*‡. * Biolojic Design LTD, ‡ Aulos Bioscience Inc.*

Abstract: Treatments based on activating effector cells against tumors are undermined by a negative feedback loop caused by endogenous IL-2 secreted from activated T effector cells. AU-007, a computationally designed human antibody, binds hIL-2 with pM affinity and completely inhibits its binding to CD25, without hindering its binding to CD122/CD132. This prevents hIL-2 from binding to the CD25/CD122/CD132 trimeric receptor expressed on Tregs, but not to the CD122/CD132 dimeric receptor expressed on Teffs and NKs. AU-007 binds to IL-2, promoting Teffs, NKs and NKTs activation while preventing Treg expansion and binding of IL-2 to vascular endothelium. AU-007 is efficacious in cancer models *in vivo*, and has an excellent safety profile in non-human primates.

Redirected IL-2 Signaling on Binding to AU-007

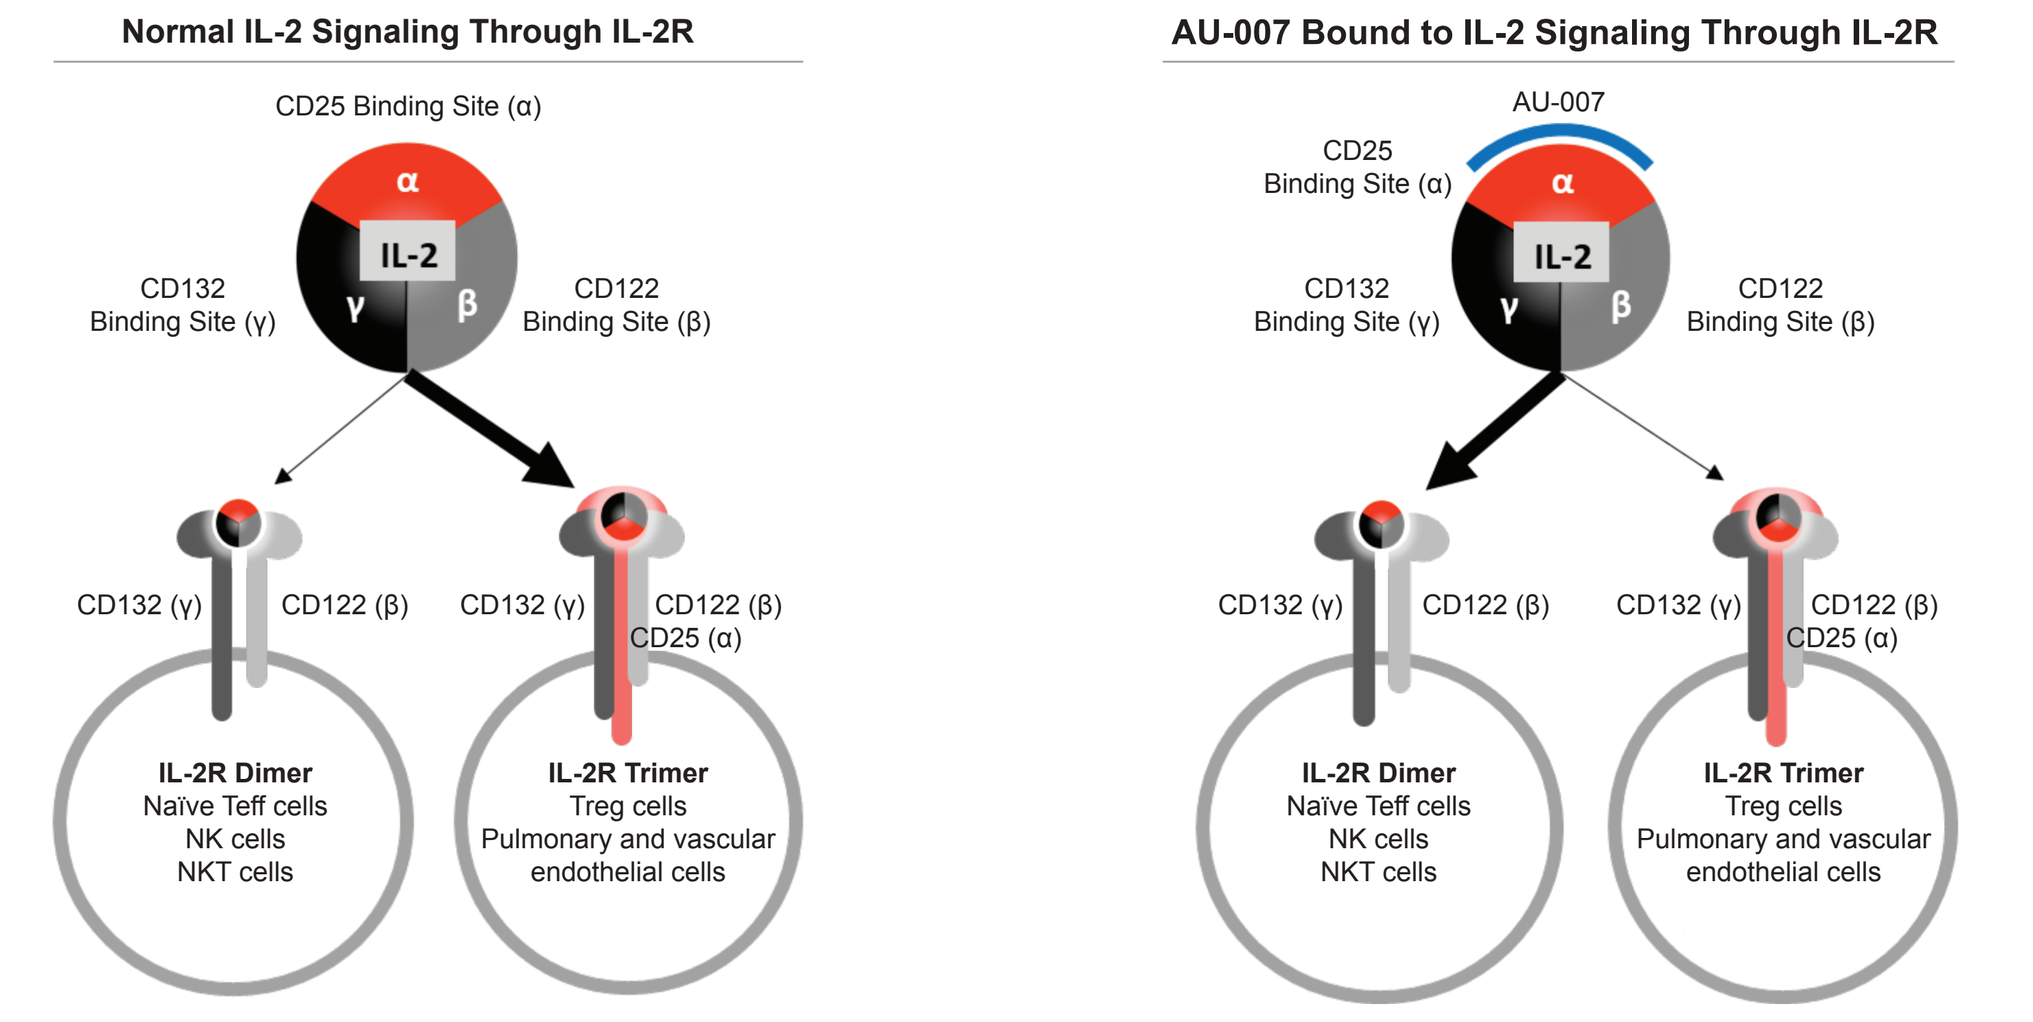
Normal IL-2 Signaling Through IL-2R
AU-007 Bound to IL-2 Signaling Through IL-2R
CD25 Binding Site (α)
CD132 Binding Site (γ)
CD122 Binding Site (β)
AU-007
IL-2
CD132 (γ)
CD122 (β)
CD25 (α)
IL-2R Dimer
Naïve Teff cells
NK cells
NKT cells
IL-2R Trimer
Treg cells
Pulmonary and vascular endothelial cells

Exogenous IL-2 Therapies and TCR Activation on Teffs Leads to Production of Endogenous IL-2

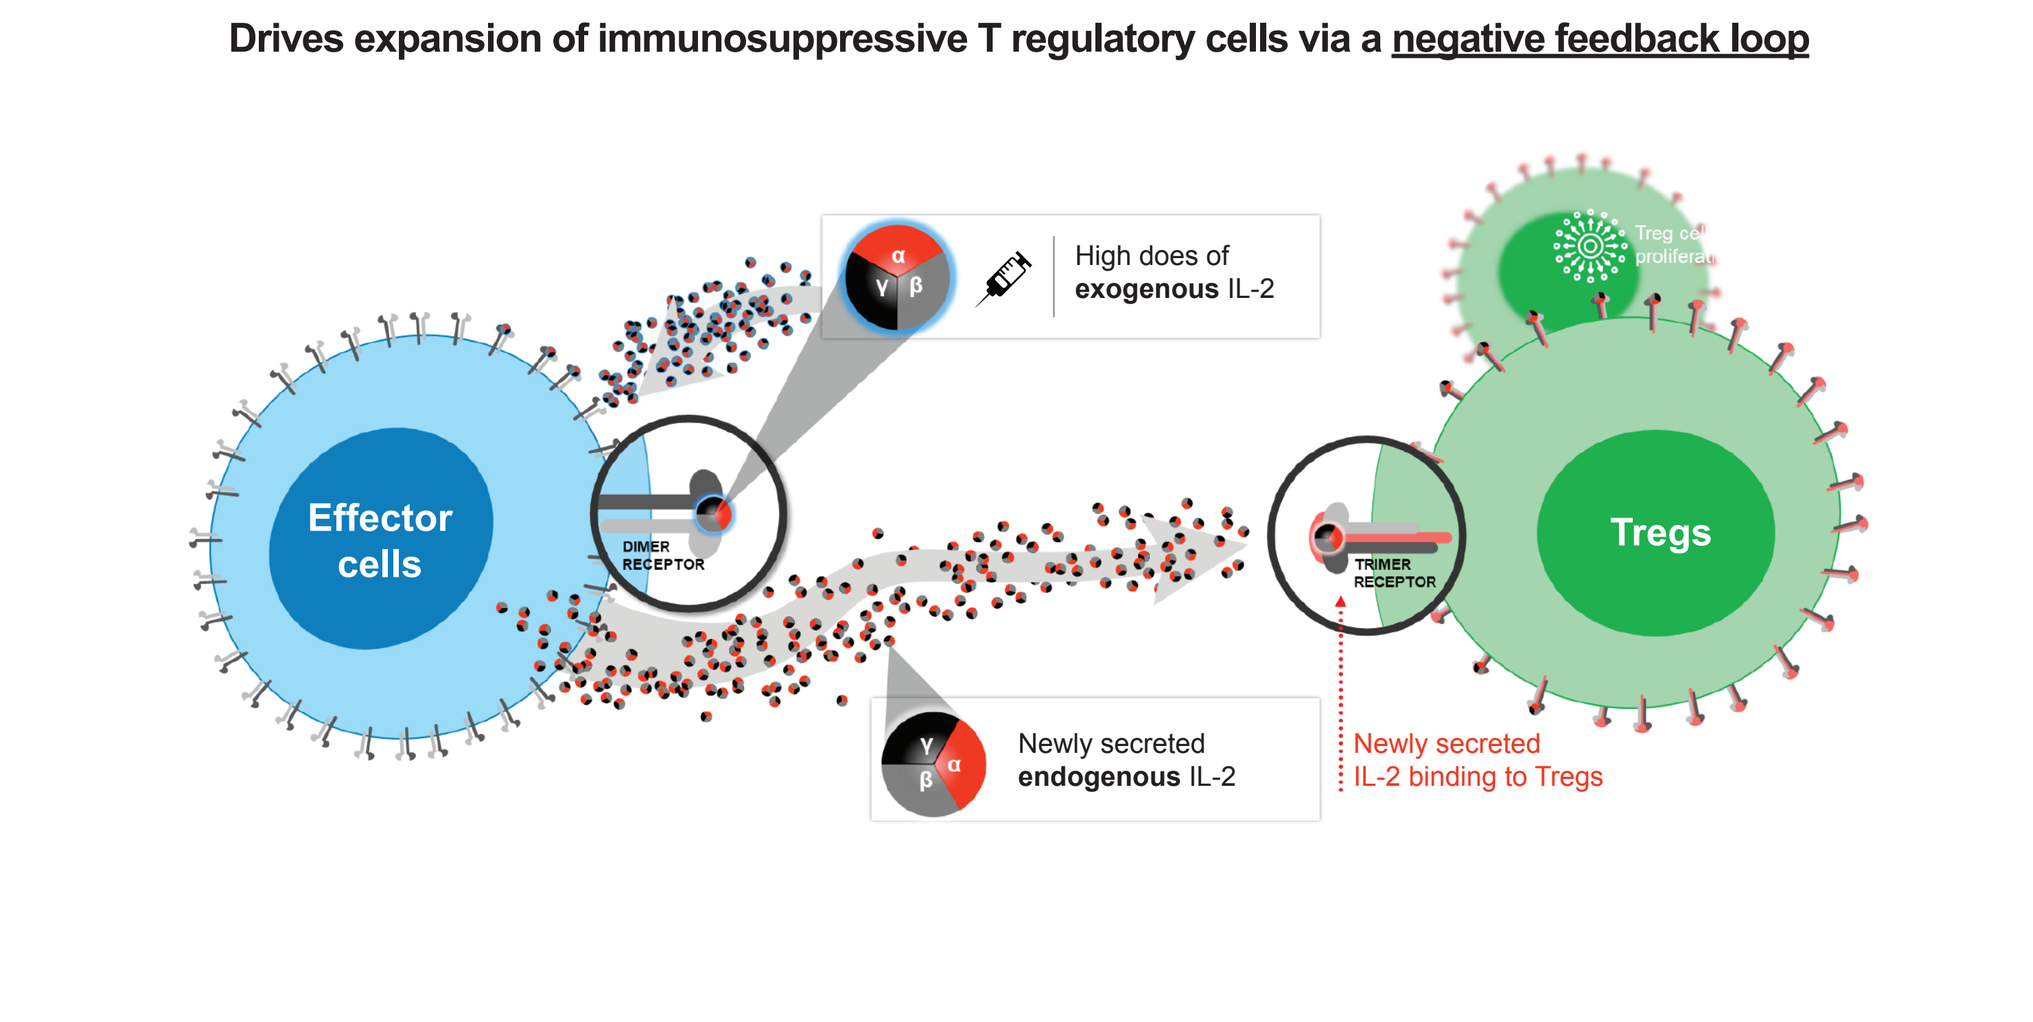
Drives expansion of immunosuppressive T regulatory cells via a negative feedback loop
High does of exogenous IL-2
Effector cells
DIMER RECEPTOR
TRIMER RECEPTOR
Tregs
Newly secreted endogenous IL-2
Newly secreted IL-2 binding to Tregs

AU-007 Uniquely Tips the Balance Toward Immune Activation, Away from Immune Suppression

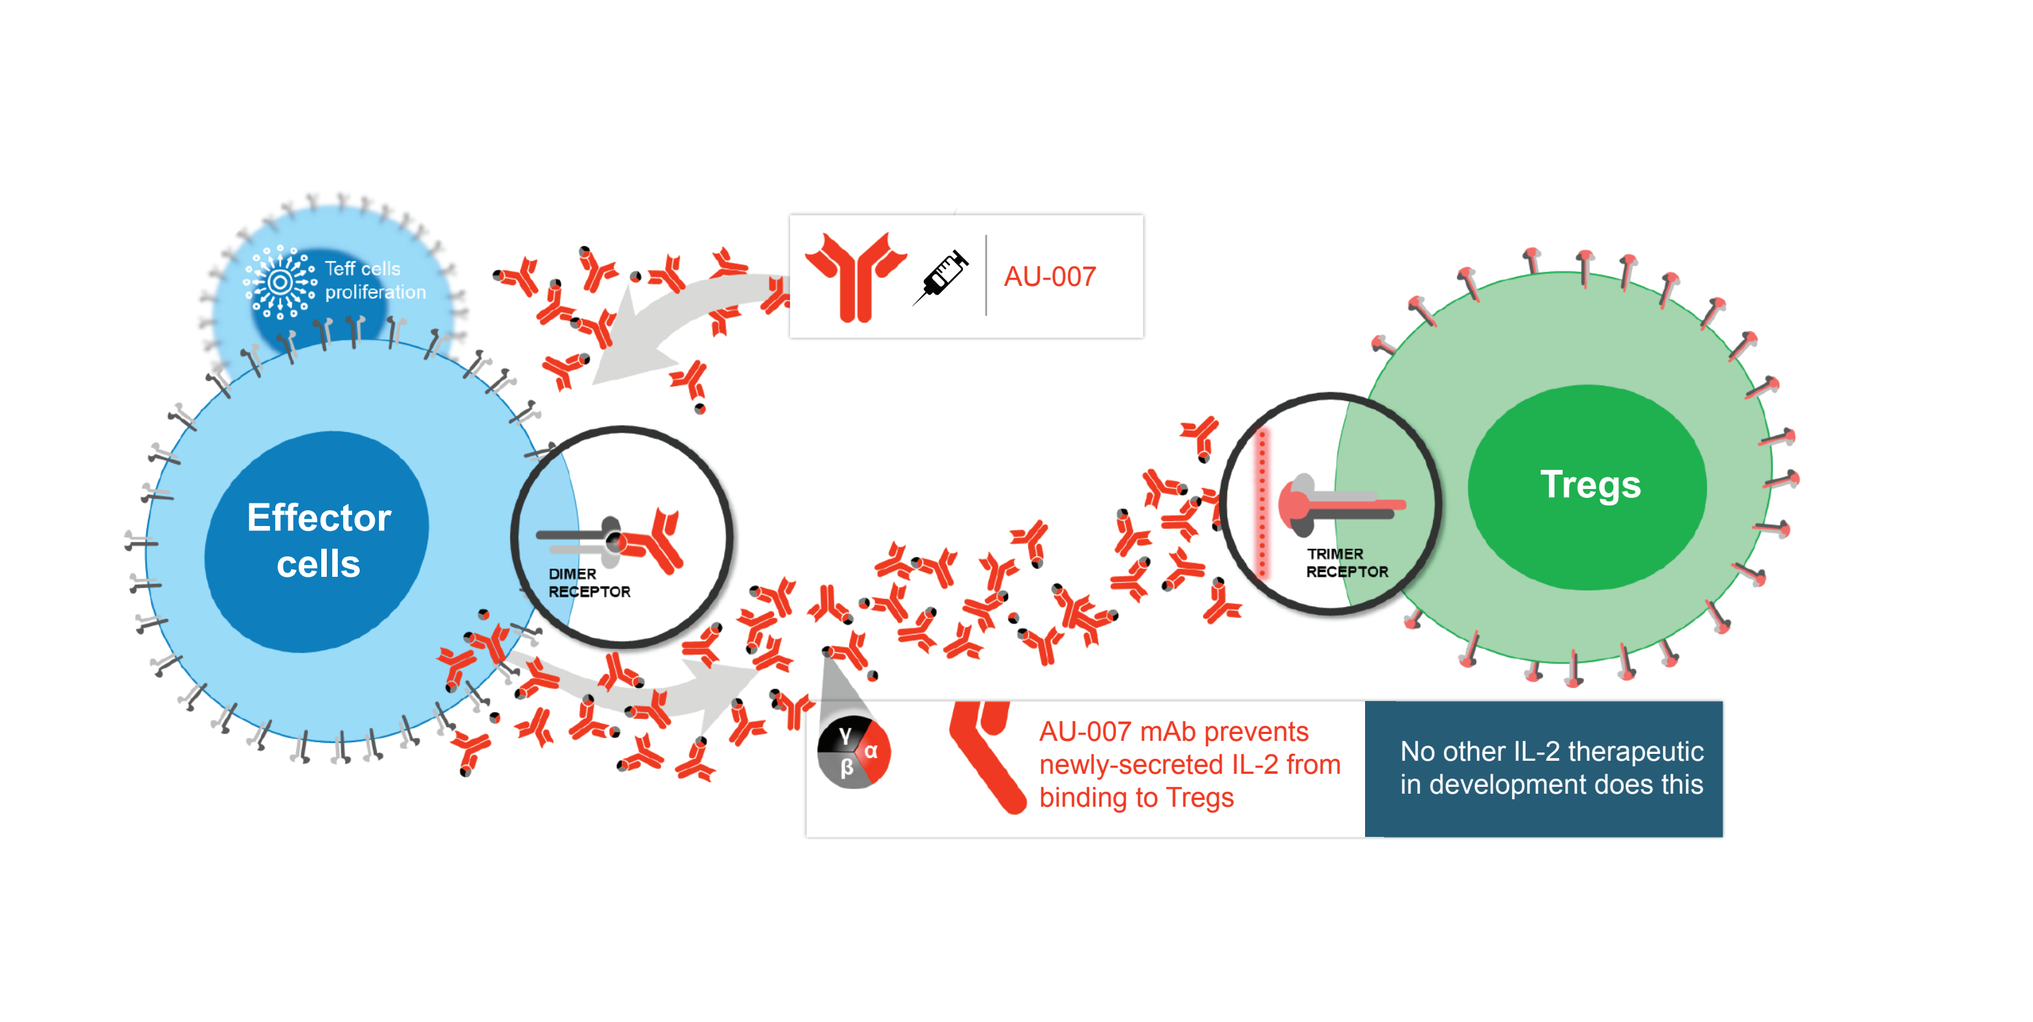
AU-007
Effector cells
Tregs
DIMER RECEPTOR
TRIMER RECEPTOR
AU-007 mAb prevents newly-secreted IL-2 from binding to Tregs
No other IL-2 therapeutic in development does this

Fig. 1. IL-2 regulation of the immune system. Breaking the negative feedback loop by AU-007 selective targeting.

AU-007 binds human IL-2 with high affinity and inhibits binding to CD25 while preserving binding to CD122

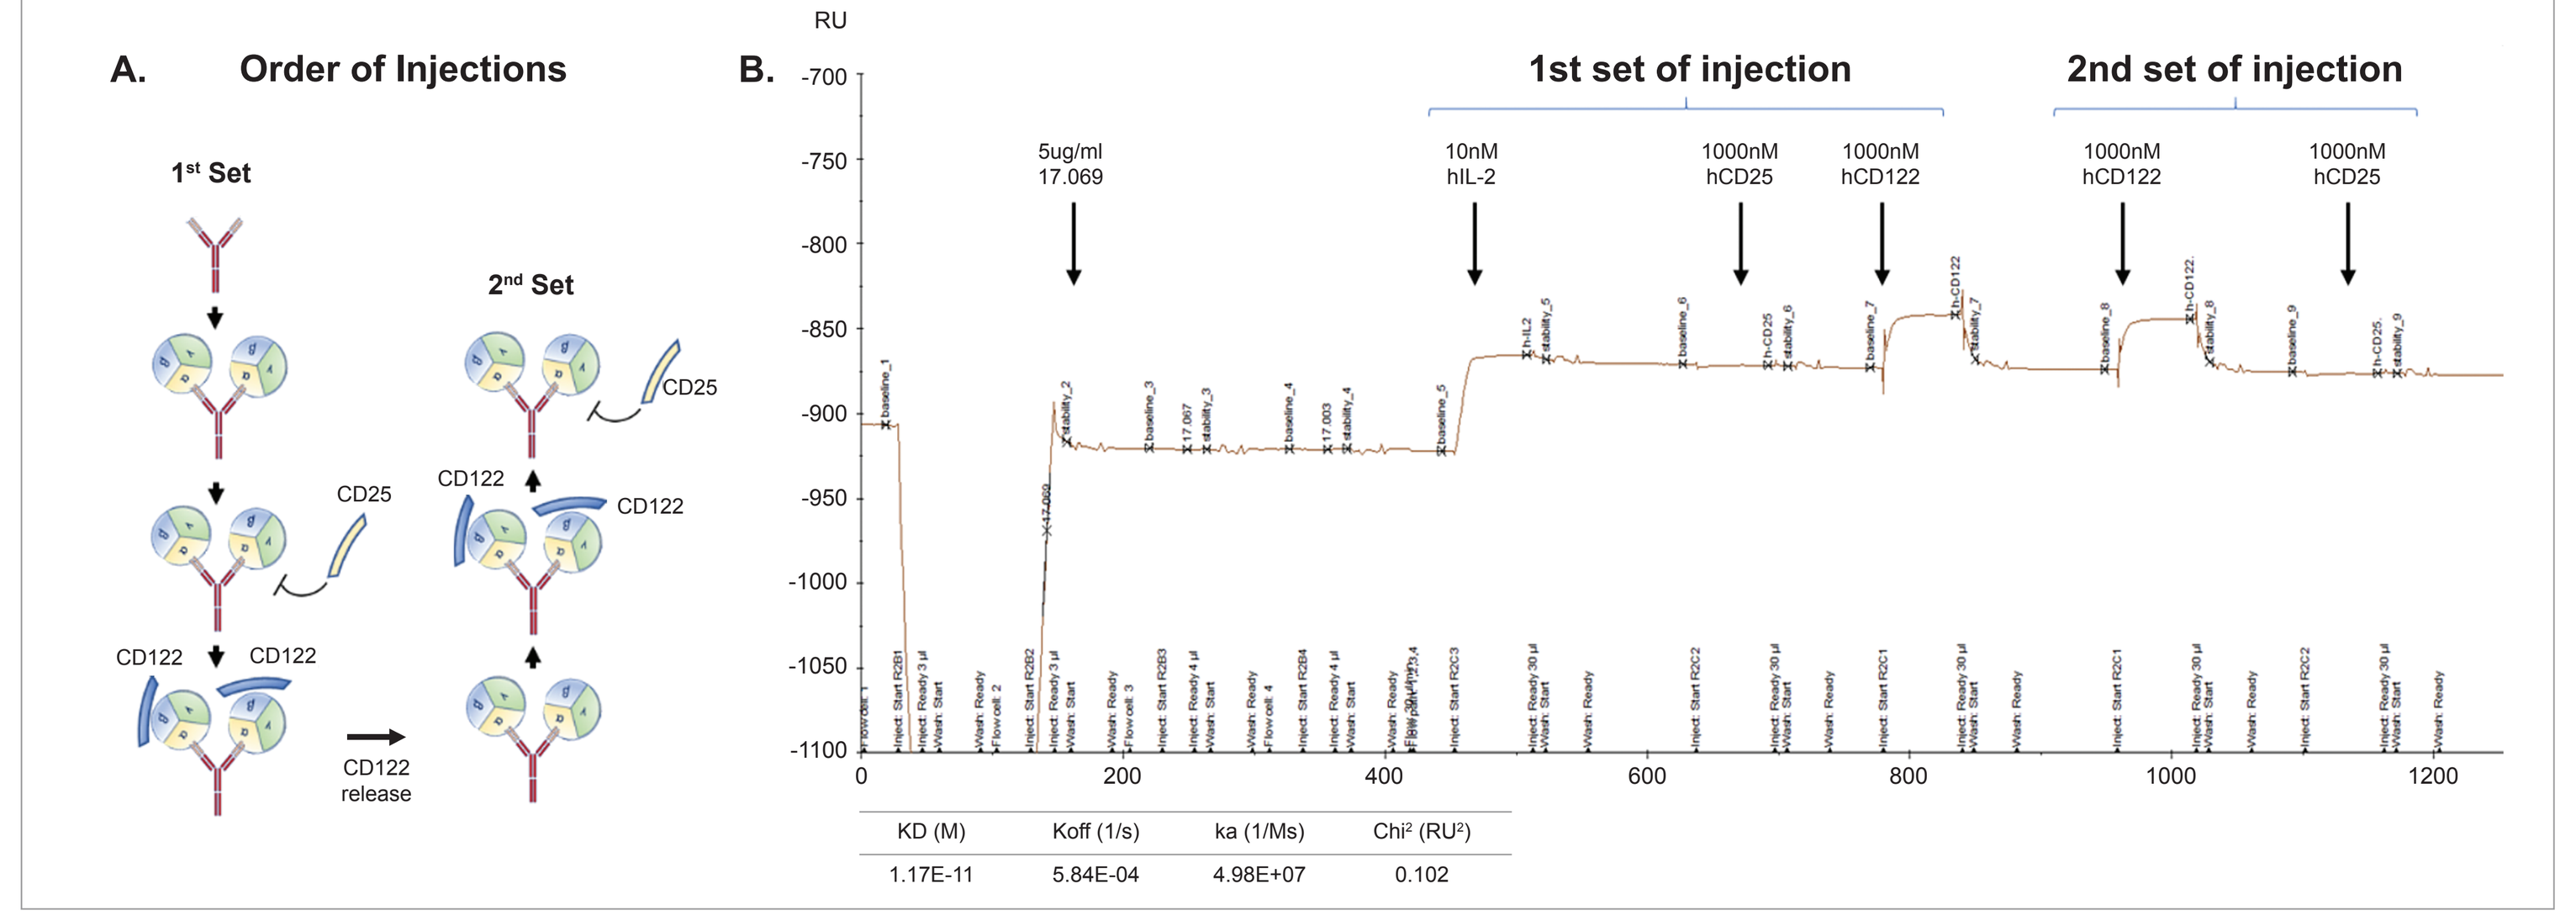
A. Order of Injections
1st Set
2nd Set
CD25
CD122
CD122 release
B.
RU
1st set of injection
2nd set of injection
5ug/ml 17.069
10nM hIL-2
1000nM hCD25
1000nM hCD122
1000nM hCD122
1000nM hCD25

KD (M)	Koff (1/s)	ka (1/Ms)	Chi² (RU²)
1.17E-11	5.84E-04	4.98E+07	0.102

Fig. 2. AU-007 binds human IL-2 with high affinity and inhibits binding to CD25 while preserving binding to CD122. Affinity and binding site are demonstrated using Surface Plasmon Resonance. AU-007 was captured on CM5 chip and soluble hIL-2 was injected, forming a complex. Subsequently, soluble CD25 was injected followed by injection of soluble CD122 (A). SPR trace of complex formation of Ab/IL-2/IL-2R arrows indicate where AU-007 (17.069), hIL-2, CD25 and CD122 were injected (B). The table presents SPR binding kinetics of AU-007 to hIL-2 with a KD of 11.7pM.

AU-007 elicits *in vivo* potent immune stimulating effects in a dose dependent manner, with no observed effect on Tregs

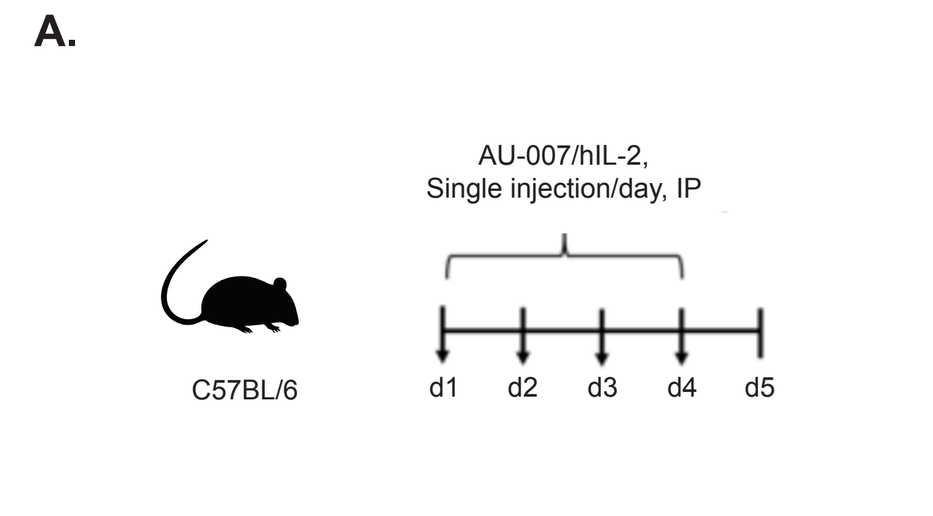
A.
AU-007/hIL-2, Single injection/day, IP
C57BL/6
d1 d2 d3 d4 d5

Fig. 3. AU-007 elicits in vivo potent immune stimulating effects in a dose dependent manner, with no observed effect on Tregs. C57BL/6 healthy mice were administered AU-007/hIL-2 complex daily for four days. On day five splenocytes were isolated and immune cells populations were analyzed using flow cytometry. **A.** Dosing regimen outline. **B-E.** Mean values of immune cells calculated as a percentage from parent population of each experimental group (n=6 per group).

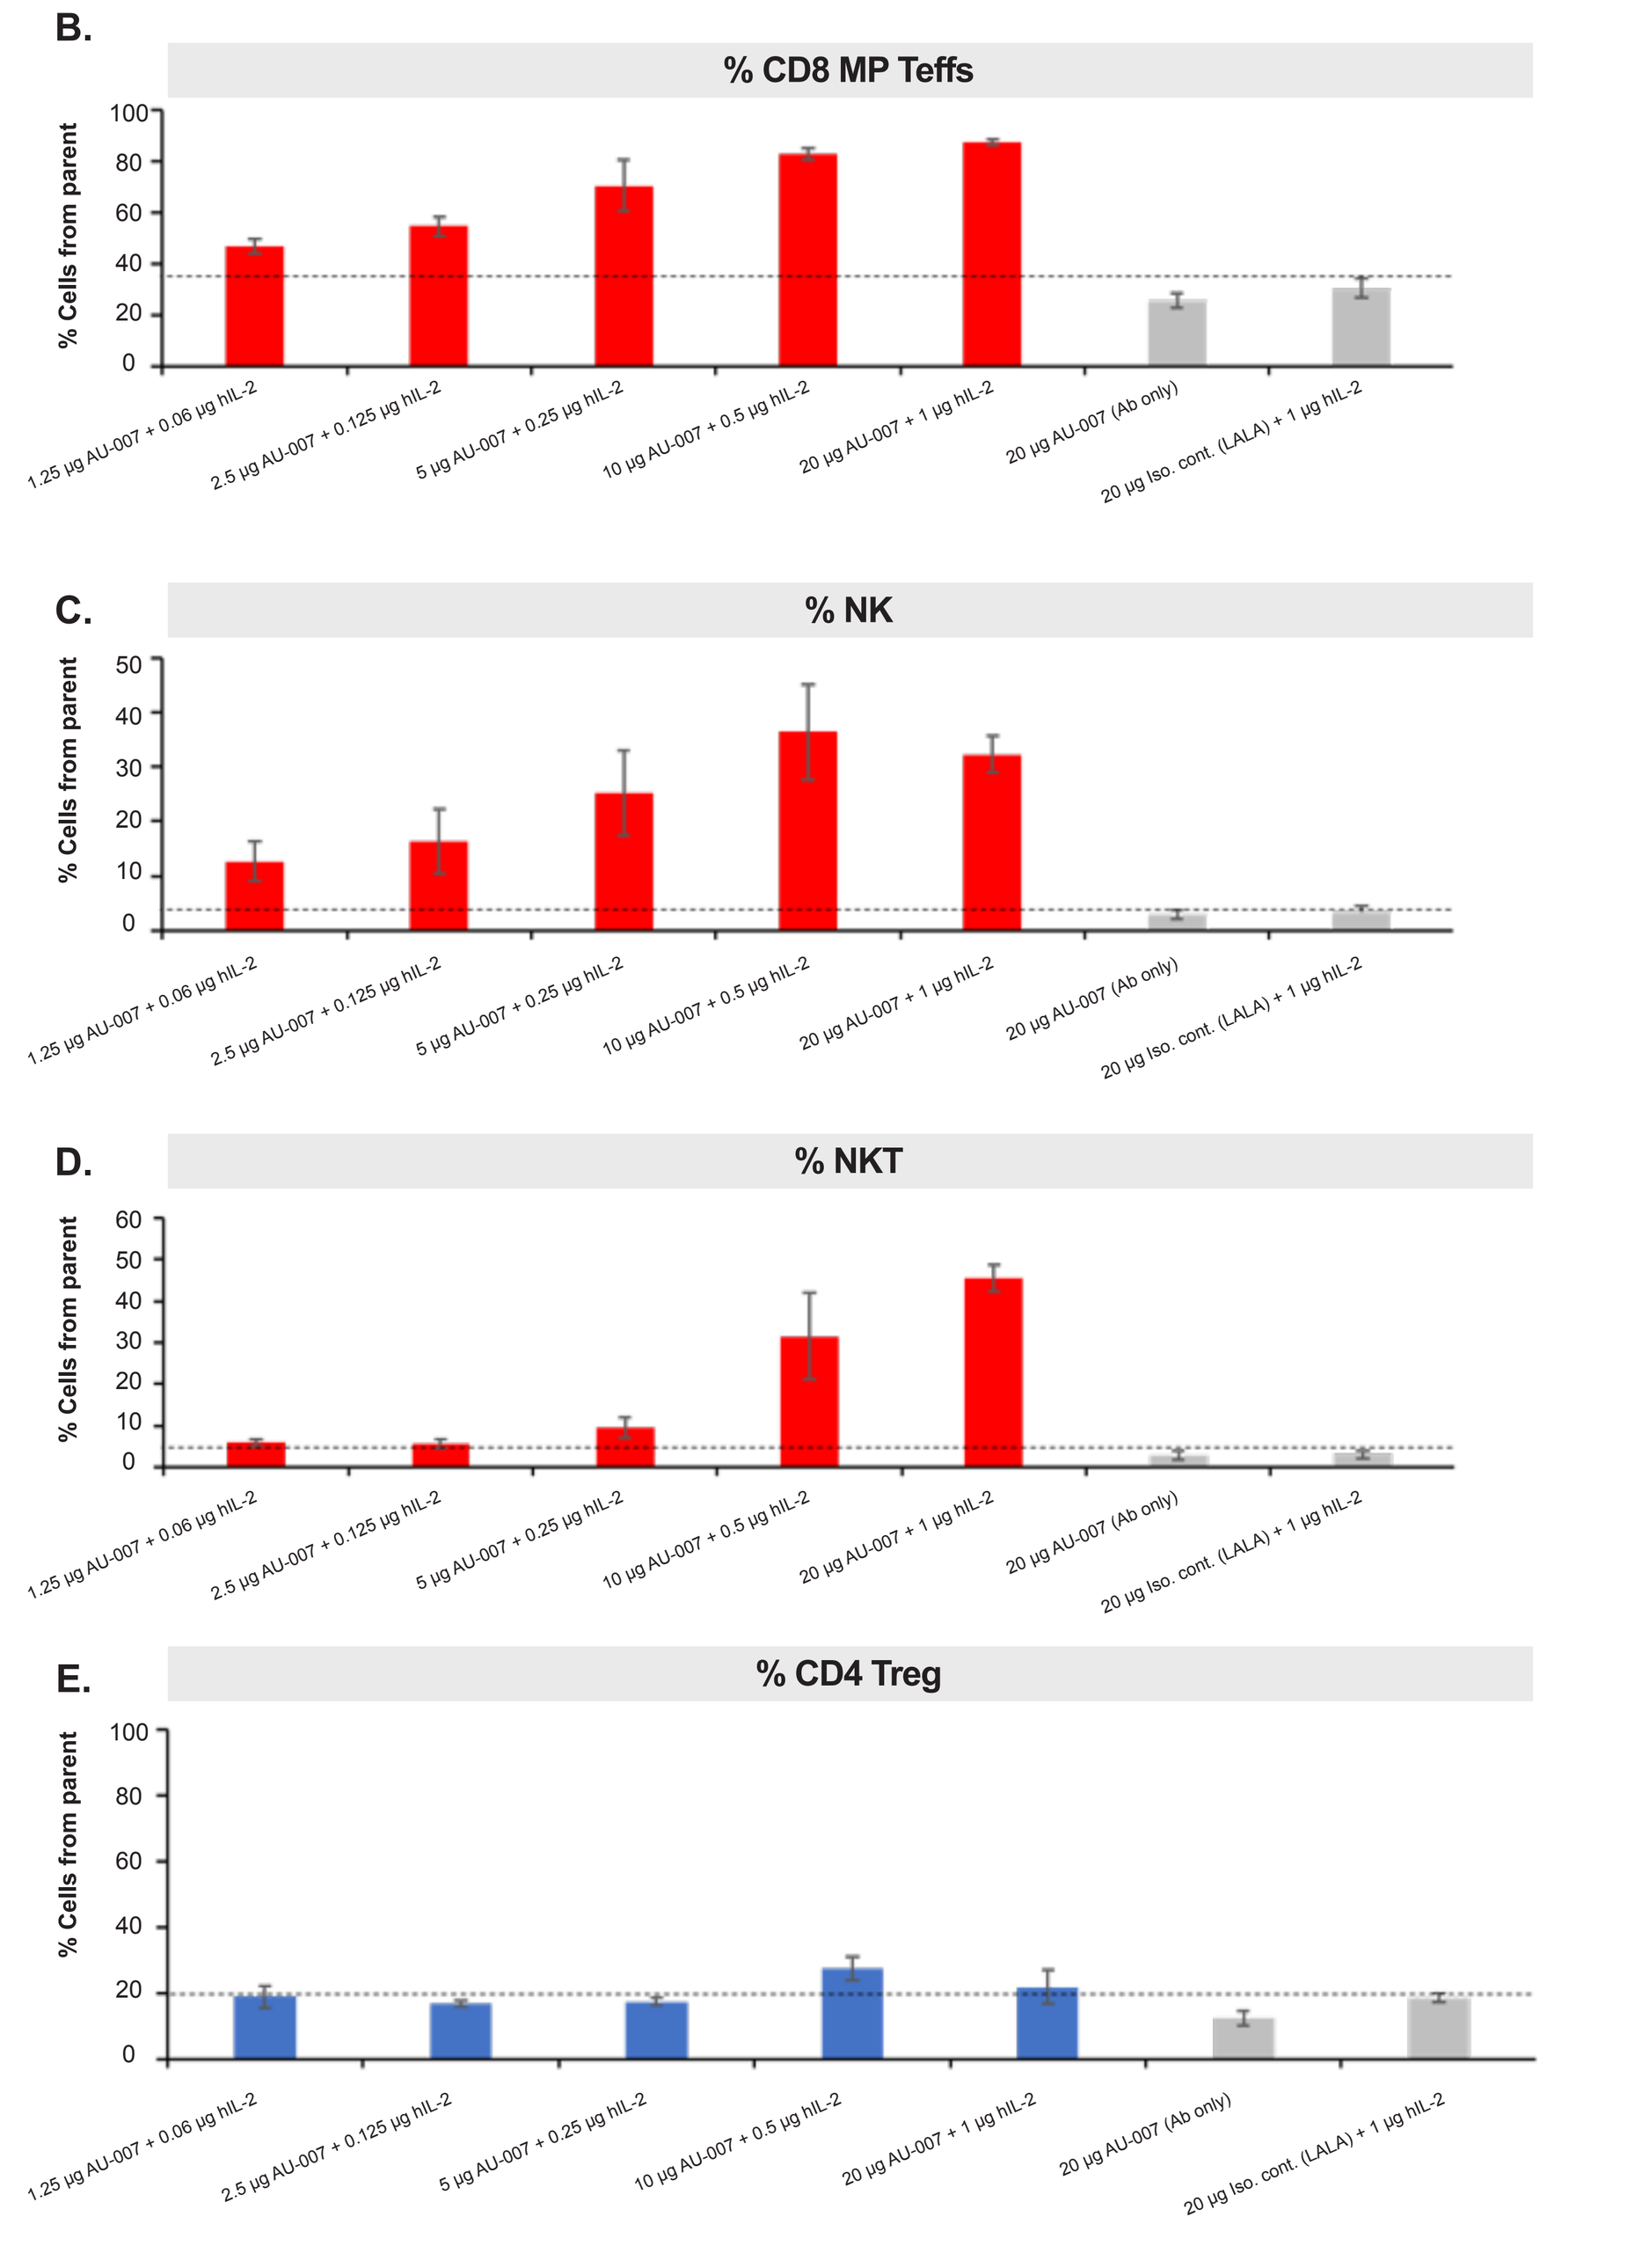
B. % CD8 MP Teffs
C. % NK
D. % NKT
E. % CD4 Treg
% Cells from parent
1.25 µg AU-007 + 0.06 µg hIL-2
2.5 µg AU-007 + 0.125 µg hIL-2
5 µg AU-007 + 0.25 µg hIL-2
10 µg AU-007 + 0.5 µg hIL-2
20 µg AU-007 + 1 µg hIL-2
20 µg AU-007 (Ab only)
20 µg Iso. cont. (LALA) + 1 µg hIL-2

AU-007 inhibits the effect of human IL-2 on Tregs while preserving its effect on Teffs, NK and NKT cells

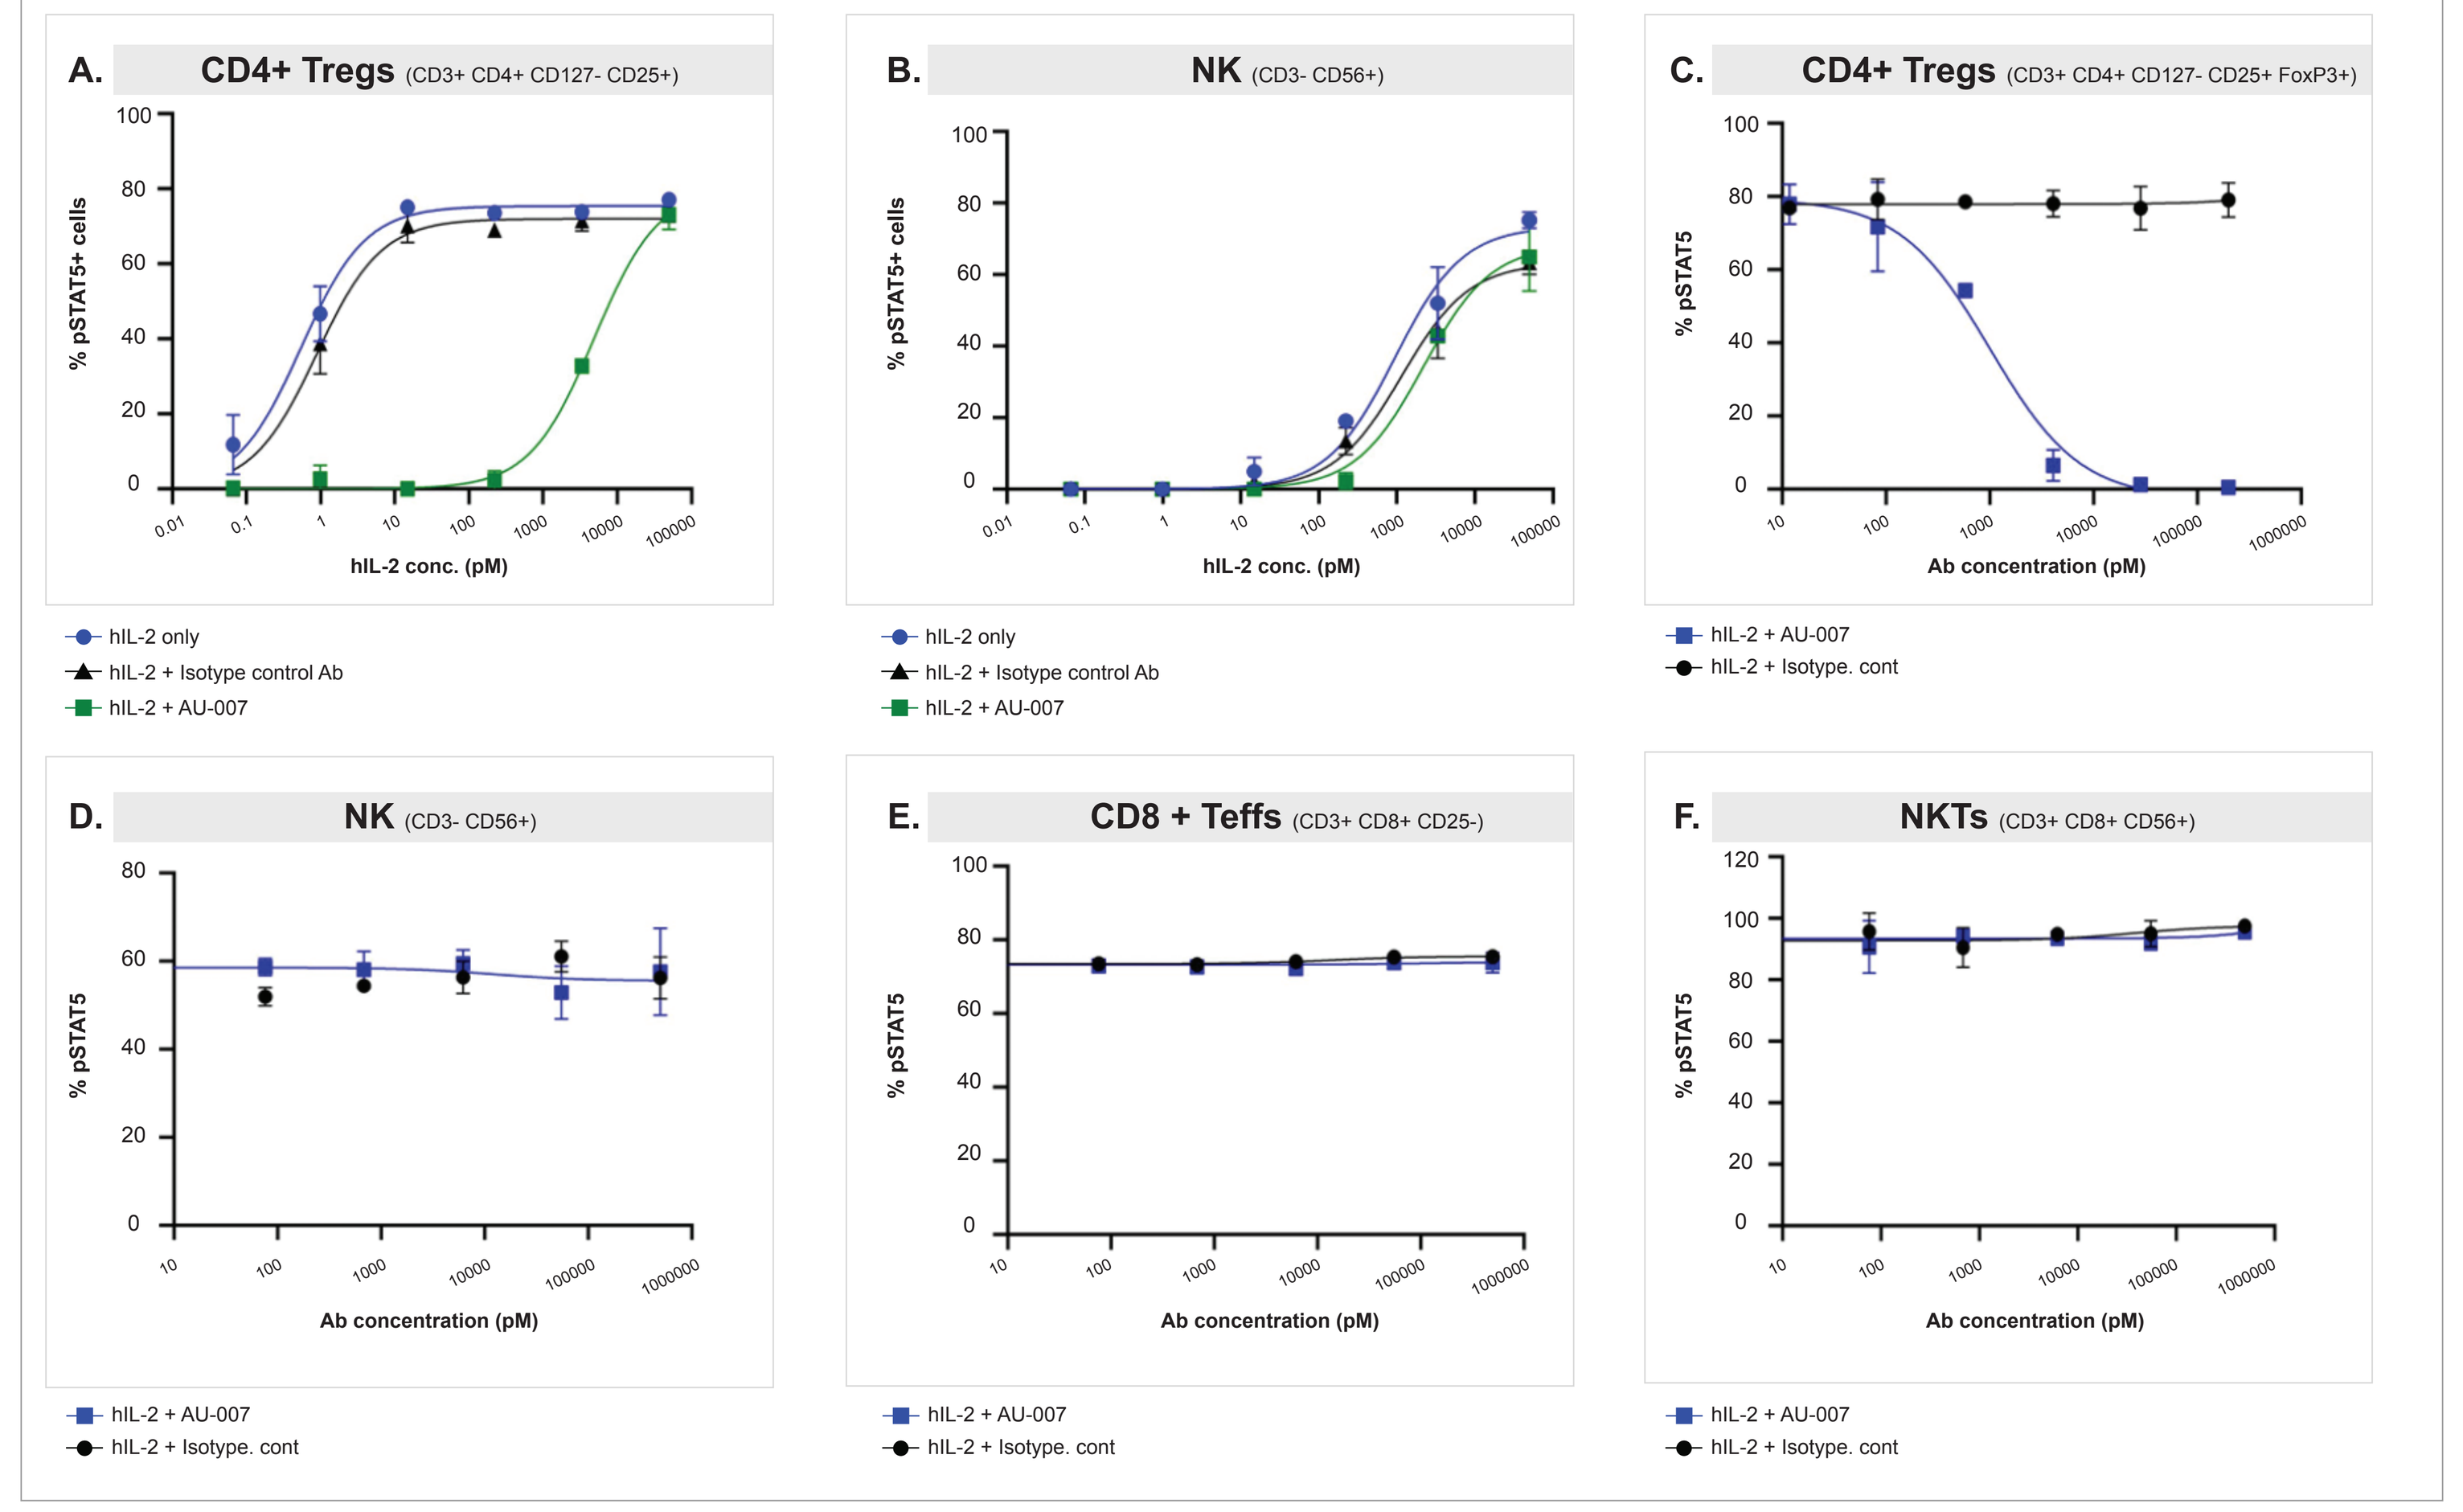
A. CD4+ Tregs (CD3+ CD4+ CD127- CD25+)
B. NK (CD3- CD56+)
C. CD4+ Tregs (CD3+ CD4+ CD127- CD25+ FoxP3+)
D. NK (CD3- CD56+)
E. CD8 + Teffs (CD3+ CD8+ CD25-)
F. NKTs (CD3+ CD8+ CD56+)
% pSTAT5+ cells
% pSTAT5
hIL-2 conc. (pM)
Ab concentration (pM)
hIL-2 only
hIL-2 + Isotype control Ab
hIL-2 + AU-007
hIL-2 + Isotype. cont

Fig. 4. AU-007 inhibits the effect of IL-2 on Tregs while preserving its effect on Teffs, NKs and NKT cells. **A-B:** Phosphorylated STAT5 levels of human immune cell subsets responding to various concentrations of hIL-2 with and without 200nM AU-007. Total naïve hPBMC culture were incubated with increasing doses of hIL-2 or with increasing doses of hIL-2 + 200nM AU-007 for 15 min. Immune cells subpopulations were analyzed by flow cytometry, gating was defined based on FMOs (fluorescence minus one control). **C-F:** Phosphorylated STAT5 levels of human immune cell subsets responding to titrated AU-007 or isotype control. Total naïve hPBMC culture were incubated with hIL-2 and with increasing doses of indicated antibody for 15 min.

AU-007 captures IL-2 and inhibits the expansion of Tregs

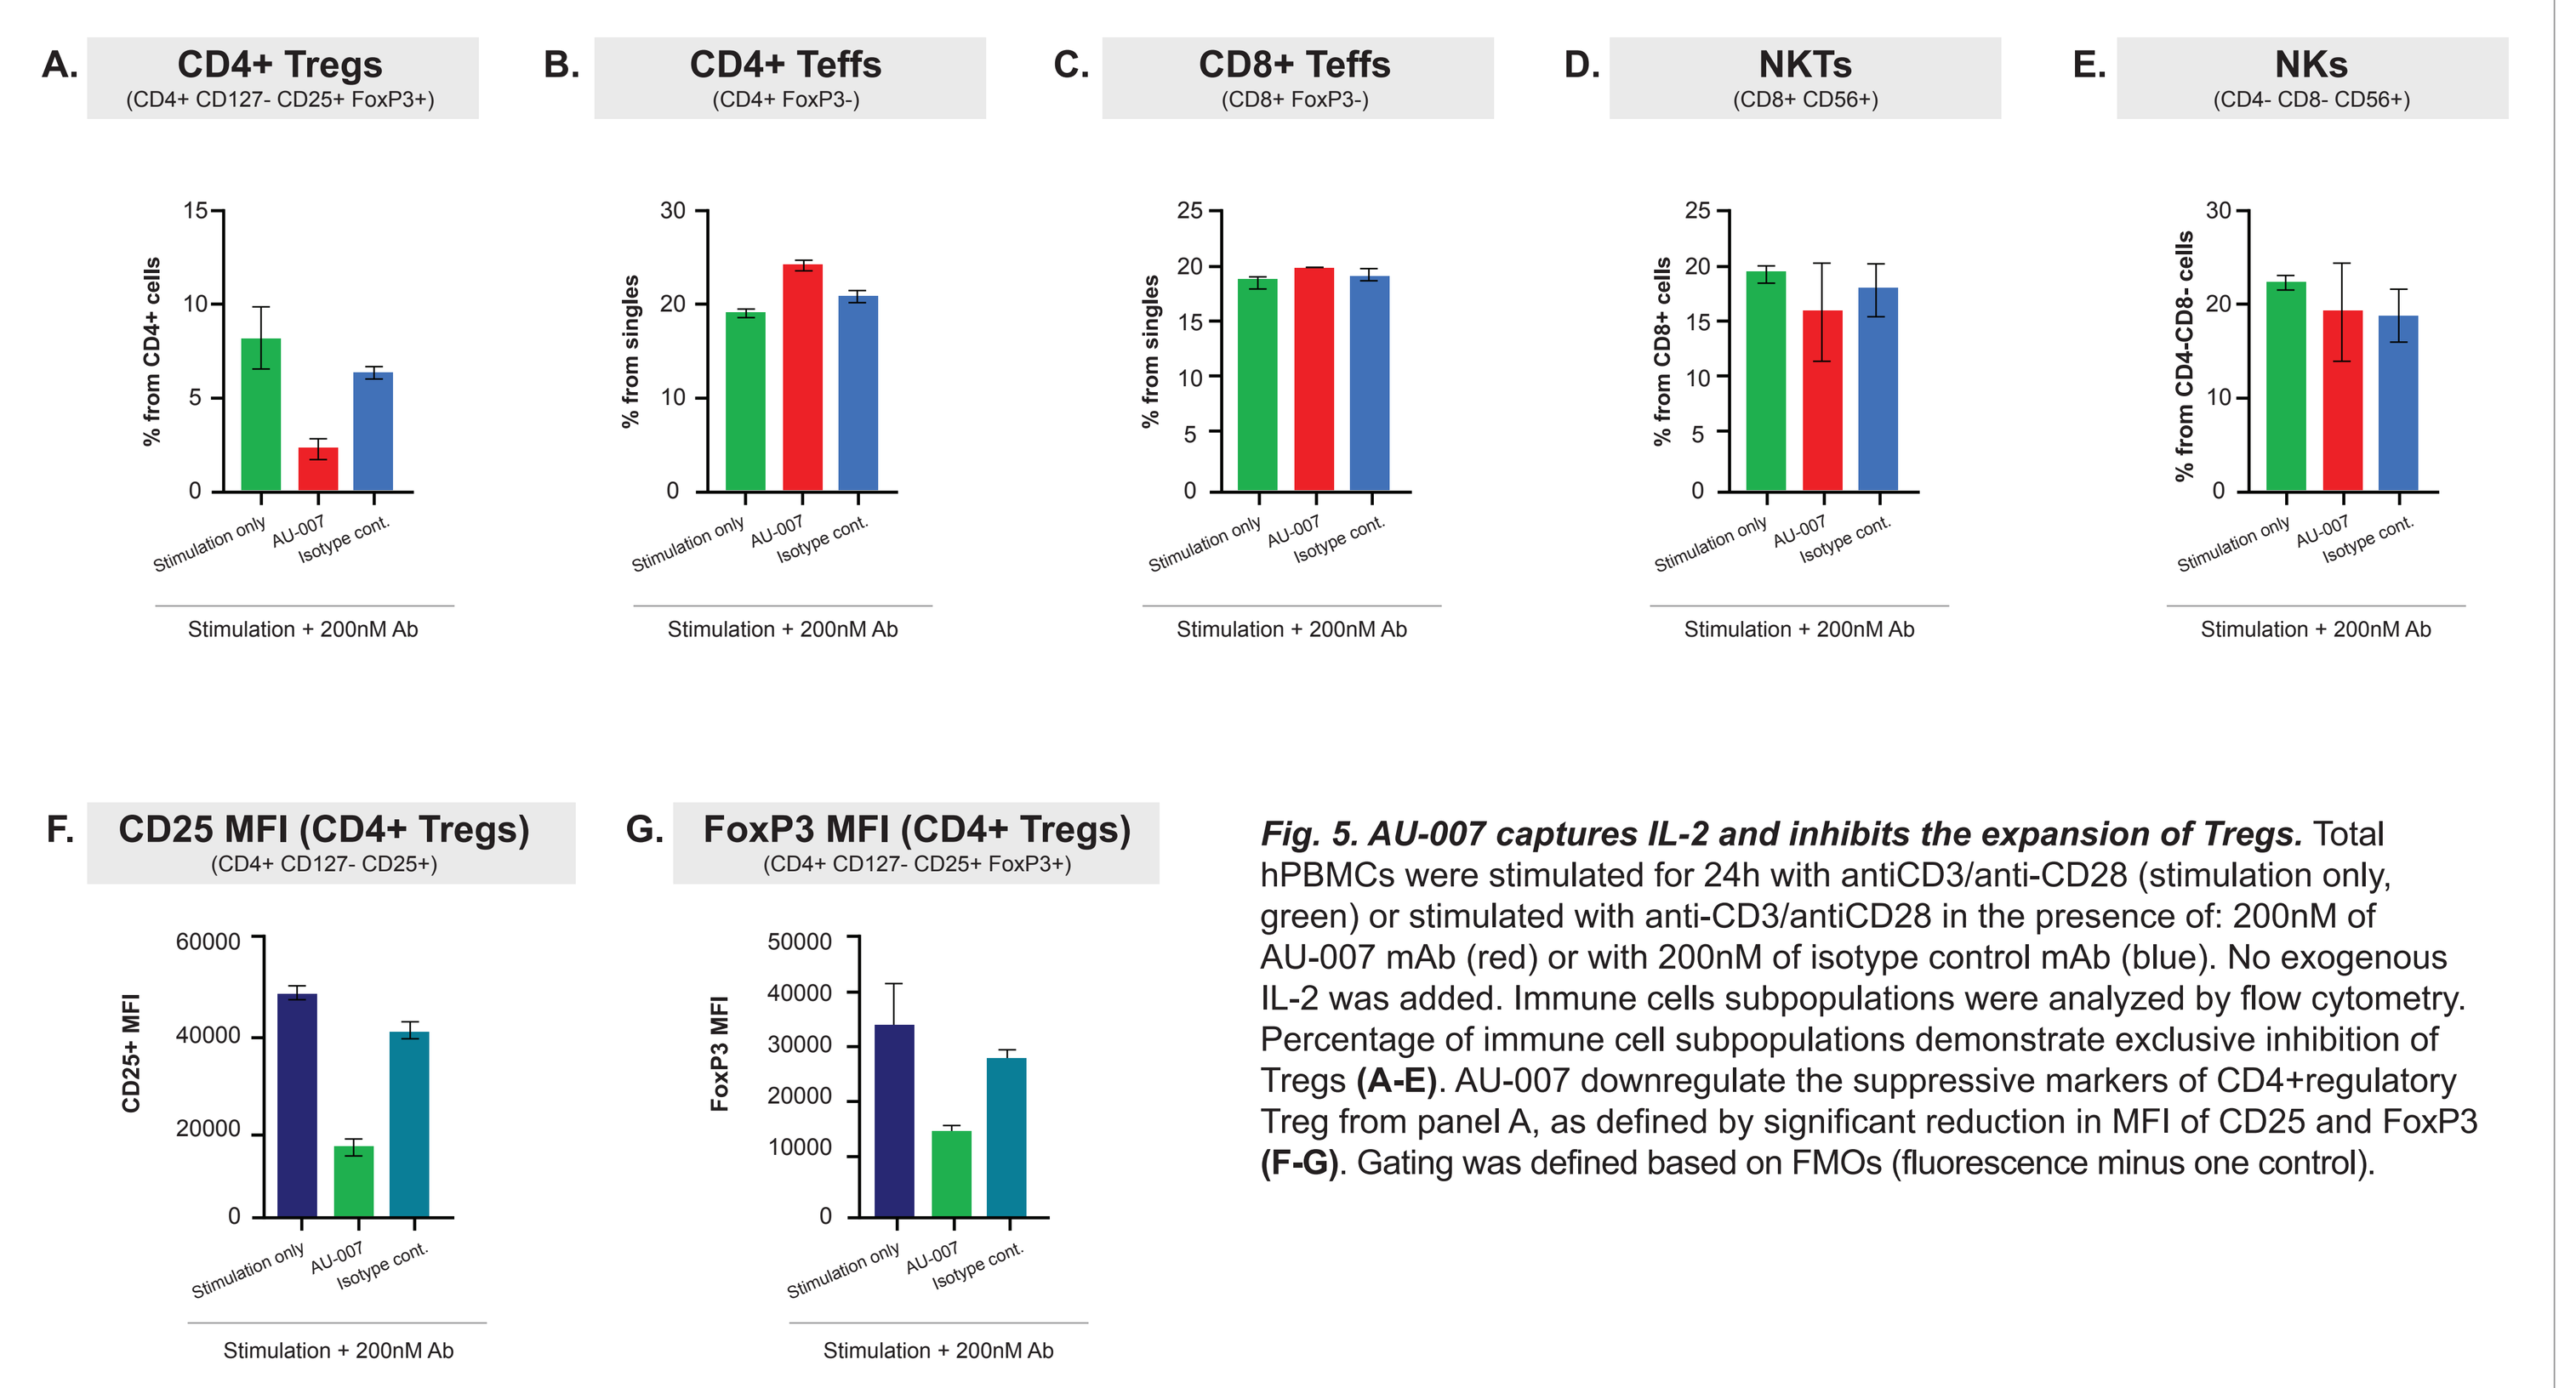
A. CD4+ Tregs (CD4+ CD127- CD25+ FoxP3+)
B. CD4+ Teffs (CD4+ FoxP3-)
C. CD8+ Teffs (CD8+ FoxP3-)
D. NKTs (CD8+ CD56+)
E. NKs (CD4- CD8- CD56+)
F. CD25 MFI (CD4+ Tregs) (CD4+ CD127- CD25+)
G. FoxP3 MFI (CD4+ Tregs) (CD4+ CD127- CD25+ FoxP3+)
Stimulation only
AU-007
Isotype cont.
Stimulation + 200nM Ab

Fig. 5. AU-007 captures IL-2 and inhibits the expansion of Tregs. Total hPBMCs were stimulated for 24h with antiCD3/anti-CD28 (stimulation only, green) or stimulated with anti-CD3/antiCD28 in the presence of: 200nM of AU-007 mAb (red) or with 200nM of isotype control mAb (blue). No exogenous IL-2 was added. Immune cells subpopulations were analyzed by flow cytometry. Percentage of immune cell subpopulations demonstrate exclusive inhibition of Tregs **(A-E)**. AU-007 downregulate the suppressive markers of CD4+regulatory Treg from panel A, as defined by significant reduction in MFI of CD25 and FoxP3 **(F-G)**. Gating was defined based on FMOs (fluorescence minus one control).

AU-007 demonstrates activity in multiple tumor models

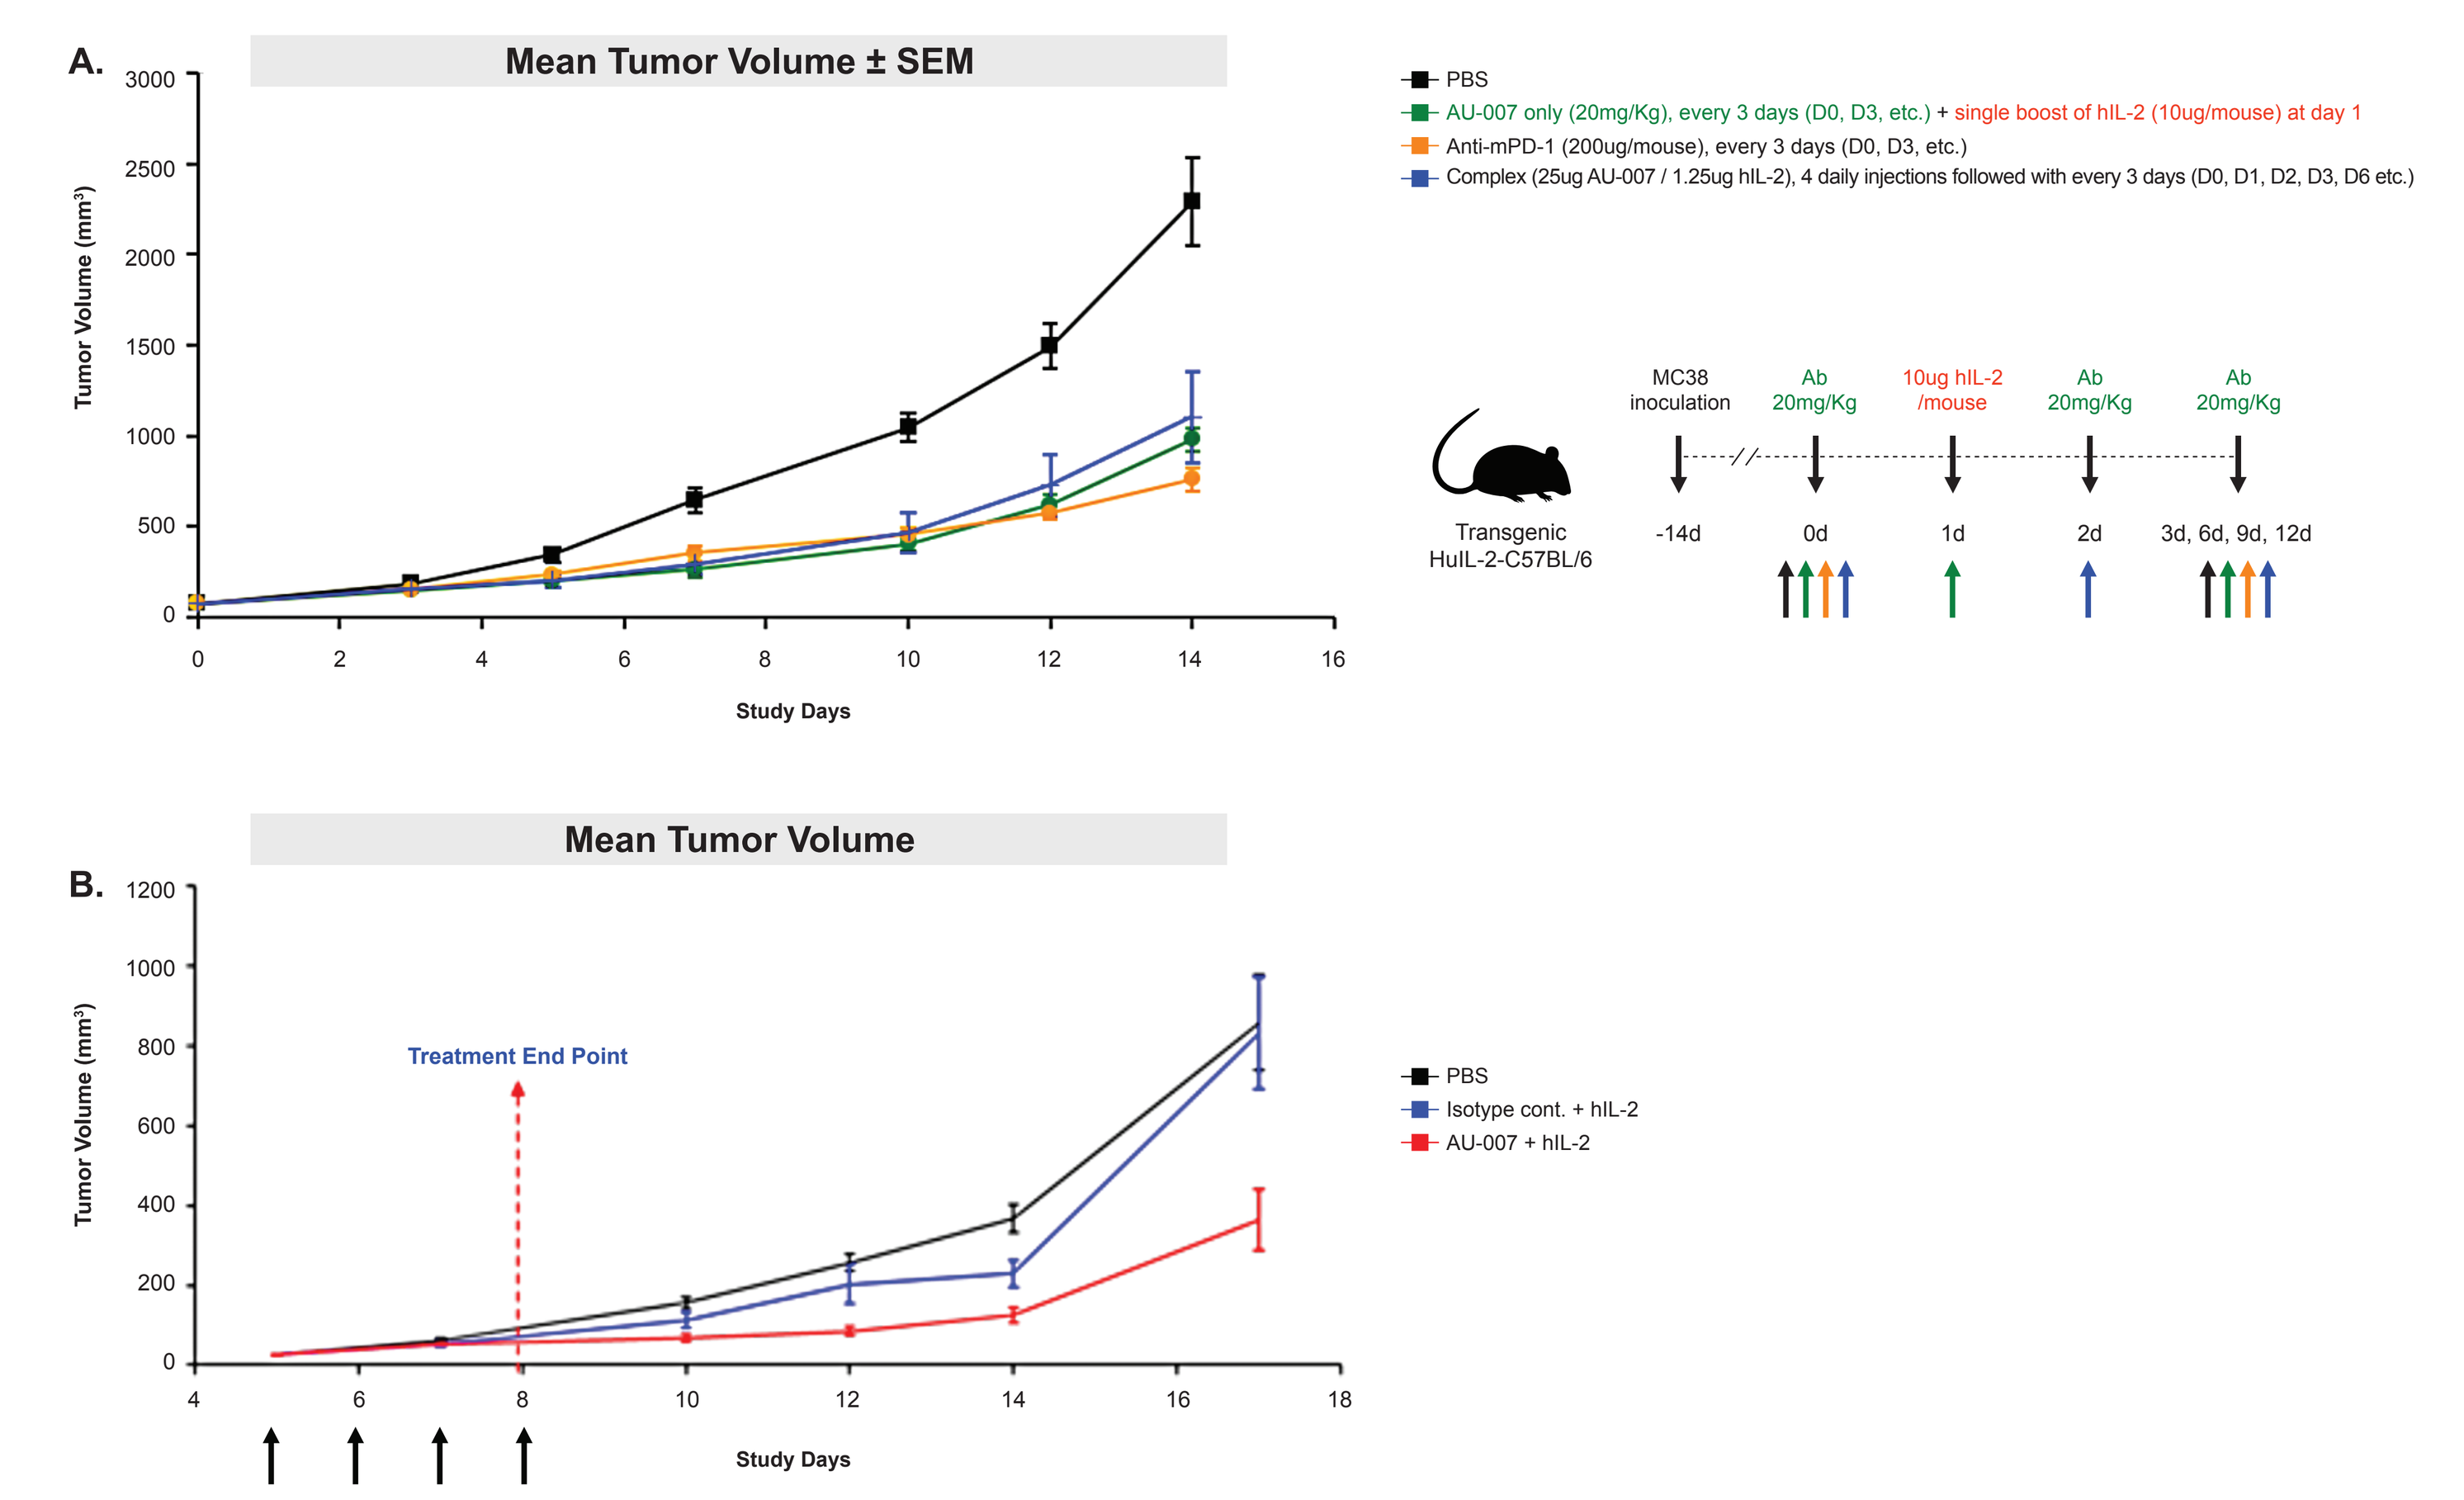
A. Mean Tumor Volume ± SEM
Tumor Volume (mm³)
Study Days
PBS
AU-007 only (20mg/Kg), every 3 days (D0, D3, etc.) + single boost of hIL-2 (10ug/mouse) at day 1
Anti-mPD-1 (200ug/mouse), every 3 days (D0, D3, etc.)
Complex (25ug AU-007 / 1.25ug hIL-2), 4 daily injections followed with every 3 days (D0, D1, D2, D3, D6 etc.)
Transgenic HuIL-2-C57BL/6
MC38 inoculation
Ab 20mg/Kg
10ug hIL-2 /mouse
-14d 0d 1d 2d 3d, 6d, 9d, 12d
B. Mean Tumor Volume
Treatment End Point
Isotype cont. + hIL-2
AU-007 + hIL-2

Fig. 6. AU-007 demonstrates activity in multiple tumor models. **A.** Genetically modified C57BL/6 mice, engineered to express human IL-2 in the background of complete knockout of mouse IL-2, were inoculated with MC38 colorectal tumor cells. All animals treated with AU-007 showed significant inhibition in tumor growth with no observed significant adverse effects Top. Administration outline: PBS (black), anti-mouse-PD-1 antibody (yellow), AU-007 pre-complexed with low dose IL-2 (blue) and AU-007 alone every three days with a single kick-starting boost of hIL-2 on day 1 (green, IL-2 single dose is marked in red). Tumor growth progression of the four groups treated and experimental outline are presented. **B.** C57BL/6 healthy mice were inoculated with B16F10 melanoma tumor cells (day 0) and administered daily, with single injection per day of Ab/hIL-2 mix (20ug/1ug respectively) or with PBS for four days. Mice were monitored daily for tumor volume. Tumor growth progression of the three groups treated is presented.

Conclusions: AU-007 is a high affinity human antibody that specifically binds to the epitope of IL-2 that interacts with CD25. These data demonstrate that AU-007 redirects IL-2 to promote Teff, NK and NKT cell expansion while blocking Treg expansion and the negative feedback loop. AU-007 demonstrates tumor growth inhibition in a transgenic mouse model that expresses human rather than murine IL-2. AU-007 is safe and well tolerated in primate toxicology studies (data not shown), and is expected to enter human clinical testing in 2021.

- Potential for higher efficacy, based on unique mechanism of action
- Potential for lower toxicity — by blocking IL-2's binding to vascular endothelium
- Unique antibody computationally designed by world-class machine learning
- Known modality; a well-behaved antibody format with drug-like properties

Access this presentation via QR code

Methods: Flow-cytometry analysis of immune-cell populations of C57BL/6 mice and hPBMCs. Tumor-Growth-Index of murine cancer models.

Presented at the 36th Annual Meeting of the Society for Immunotherapy of Cancer (SITC), November 10-14, 2021